Targeted Tumor-Derived Cellular Immunotherapy in Advanced Breast Cancer Patients Induces Initial Immune Responses and Tumor Regression.

Vivekananda (Vivek) Sunkari[1], William V. Williams[1], George E. Peoples[2], Sanne Graeve[1], Charles L. Wiseman[1], and Markus D. Lacher[1]

[1]BriaCell Therapeutics Corp., Berkeley, CA, USA; [2]Cancer Insight, LLC; San Antonio, TX, USA

ABSTRACT

Background: SV-BR-1-GM (Bria-IMT) is a GM-CSF-engineered breast cancer cell line that expresses HER2, the cancer/testis antigen PRAME, and Class I and Class II HLA antigens. Regression of metastatic breast cancer was seen in clinical trials using irradiated SV-BR-1-GM as a targeted immunotherapy. This is likely attributable to the potentially unique mechanism of action of SV-BR-1-GM.

Methods: Although derived from a breast cancer cell line, SV-BR-1-GM also resembles dendritic cells and as such may effectively activate breast cancer antigen-specific T cells (*Front Immunol.* 2018; 9:776). In a phase I/IIa trial of Bria-IMT™ in patients with advanced breast cancer, 23 patients have been inoculated with an SV-BR-1-GM regimen including low-dose cyclophosphamide to reduce immune suppression and local interferon-α2b to boost the response.

Results: Confirming previous work, several patients showed tumor regressions. Interestingly, all patients with tumor regression allele-matched SV-BR-1-GM at ≥ 1 HLA locus. Analysis of immunologic factors important for clinical outcomes such as delayed-type hypersensitivity and antibody responses suggesting heightened immune activity in patients treated with Bria-IMT™. HLA-matching subjects had an improved rate of tumor regression and also correlated with decreases in circulating cancer-associated macrophage-like cells.

Conclusions: HLA matching is currently being evaluated as a predictor of the tumor response in the ongoing combination study of Bria-IMT™ with KEYTRUDA® [manufactured by Merck & Co., Inc. (NYSE: MRK)], listed in ClinicalTrials.gov as NCT03328026. These data support the Company's Bria-OTS™ program, which is genetically modifying the SV-BR-1 cell line to express different HLA types.

(*):(edited from submitted abstract – here new data is included from Combination clinical trial of SV-BR-1-GM and KEYTRUDA®)

BACKGROUND & OBJECTIVES

1. SV-BR-1-GM is a breast cancer cell line with features of antigen-presenting cells including expression of Class II HLA molecules.
2. Prior investigation indicates that patients who match SV-BR-1-GM at 1 or more HLA alleles may be more likely to respond to treatment
3. Study-1: The SV-BR-1-GM regimen includes: low dose cyclophosphamide to reduce immune suppression (300 mg/m^2 2-3 days prior to inoculation); 20-50 million irradiated SV-BR-1-GM cells intradermally; and interferon-α2b (10,000 IU x 4) 2 & 4 days later into the SV-BR-1-GM inoculation sites with cycles are every 2 weeks x3 then monthly.
4. Study-2: Pembrolizumab (200 mg IV) in combination with the regimen from Study 1 with cycles every 3 weeks
5. The objective is to evaluate immune responses from subjects treated with the SV-BR-1-GM regimen alone or in combination with pembrolizumab
6. *BriaDX™ (Companion Diagnostic):* next-generation sequencing will be employed to determine the HLA types from patient samples

RESULTS

SV-BR-1-GM Proof-of-Concept Phase I (2004-2006):

- 4 patients were treated with the SV-BR-1-GM regimen
- Subject A002 presented with metastatic breast cancer. She initially responded to chemotherapy, but then relapsed with tumor spread to the breast, lungs, soft tissues and bone
- She was treated with the SV-BR-1-GM regimen and had a robust response with substantial tumor regression in the breast and bone, and complete clearance in the lungs and soft tissues
- **Out of 4 evaluable subject, A002 was the only patient who matched SV-BR-1-GM at the HLA-DRB3 locus**

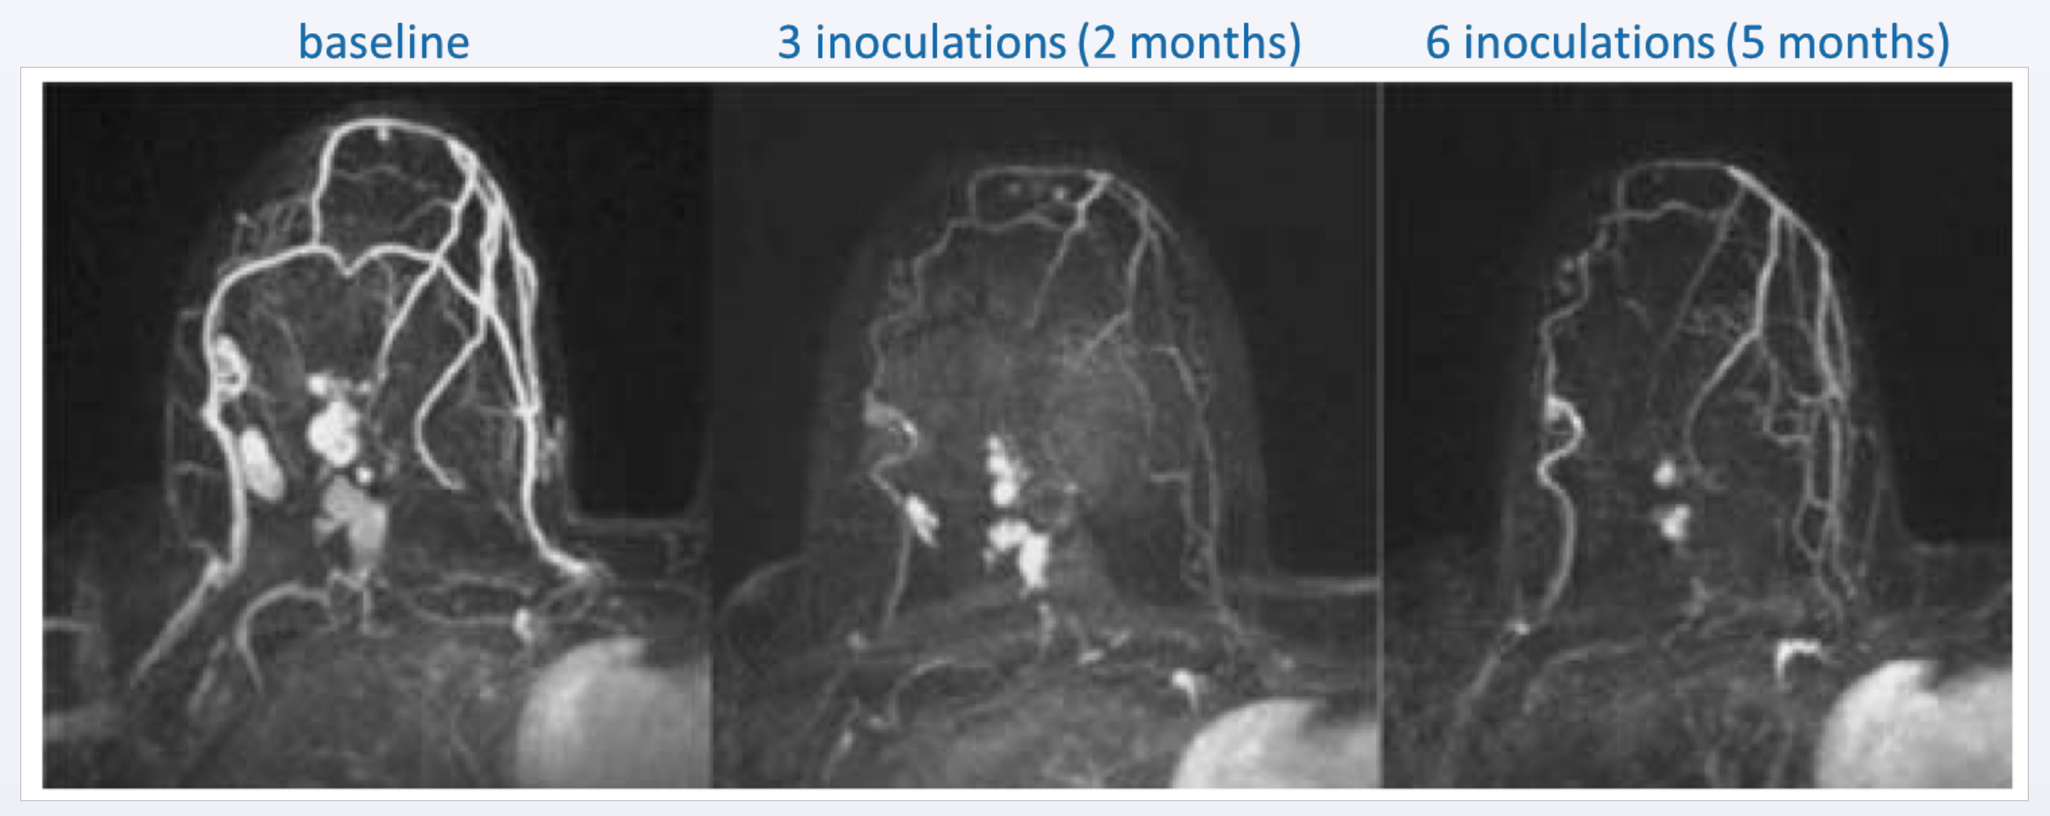
baseline
3 inoculations (2 months)
6 inoculations (5 months)

Figure 1. Tumor regression in the breast (Patient A002).
Wiseman and Kharazi, 2006.

Conclusion: Patient A002 had substantial clinical benefit from the SV-BR-1-GM regimen and matched SV-BR-1-GM at HLA-DRB3

RESULTS – Study 1 "Monotherapy"

Additional Clinical Testing in metastatic and locally recurrent breast cancer (2018-current):

- 23 subjects dosed with **SV-BR-1-GM** in phase I/IIa trial (ClinicalTrials.gov NCT03066947**). Study closed for enrollment.**

HLA Matching and Biological Activity

Patients (n)	HLA Match	Tumor Shrinkage	Biological Response*
5	≥2	40%	60%
18	≥1	22%	39%
9	0	0%	0%

*Biological response includes tumor shrinkage or lower circulating cancer associated cells

Subjects from the recent study using the SV-BR-1-GM regimen (n=23) and the evaluable patients treated in the original SV-BR-1-GM clinical trial (n=4) were evaluated for clinical tumor shrinkage and biological response based on HLA matching to SV-BR-1-GM.

Conclusion: Patients with 1+ HLA allele matches are more likely to derive clinical benefit than patients with no matches. Patients with 2+ matches are more likely to derive clinical benefit than patients with 1 match or no matches

Delayed-type hypersensitivity (DTH) is an important marker of cellular (T cell) immune responses. The largest response seen for each patient (A) and the average of all patient responses (B) are shown in SV-BR-1-GM treated patients measured by average erythema and induration.

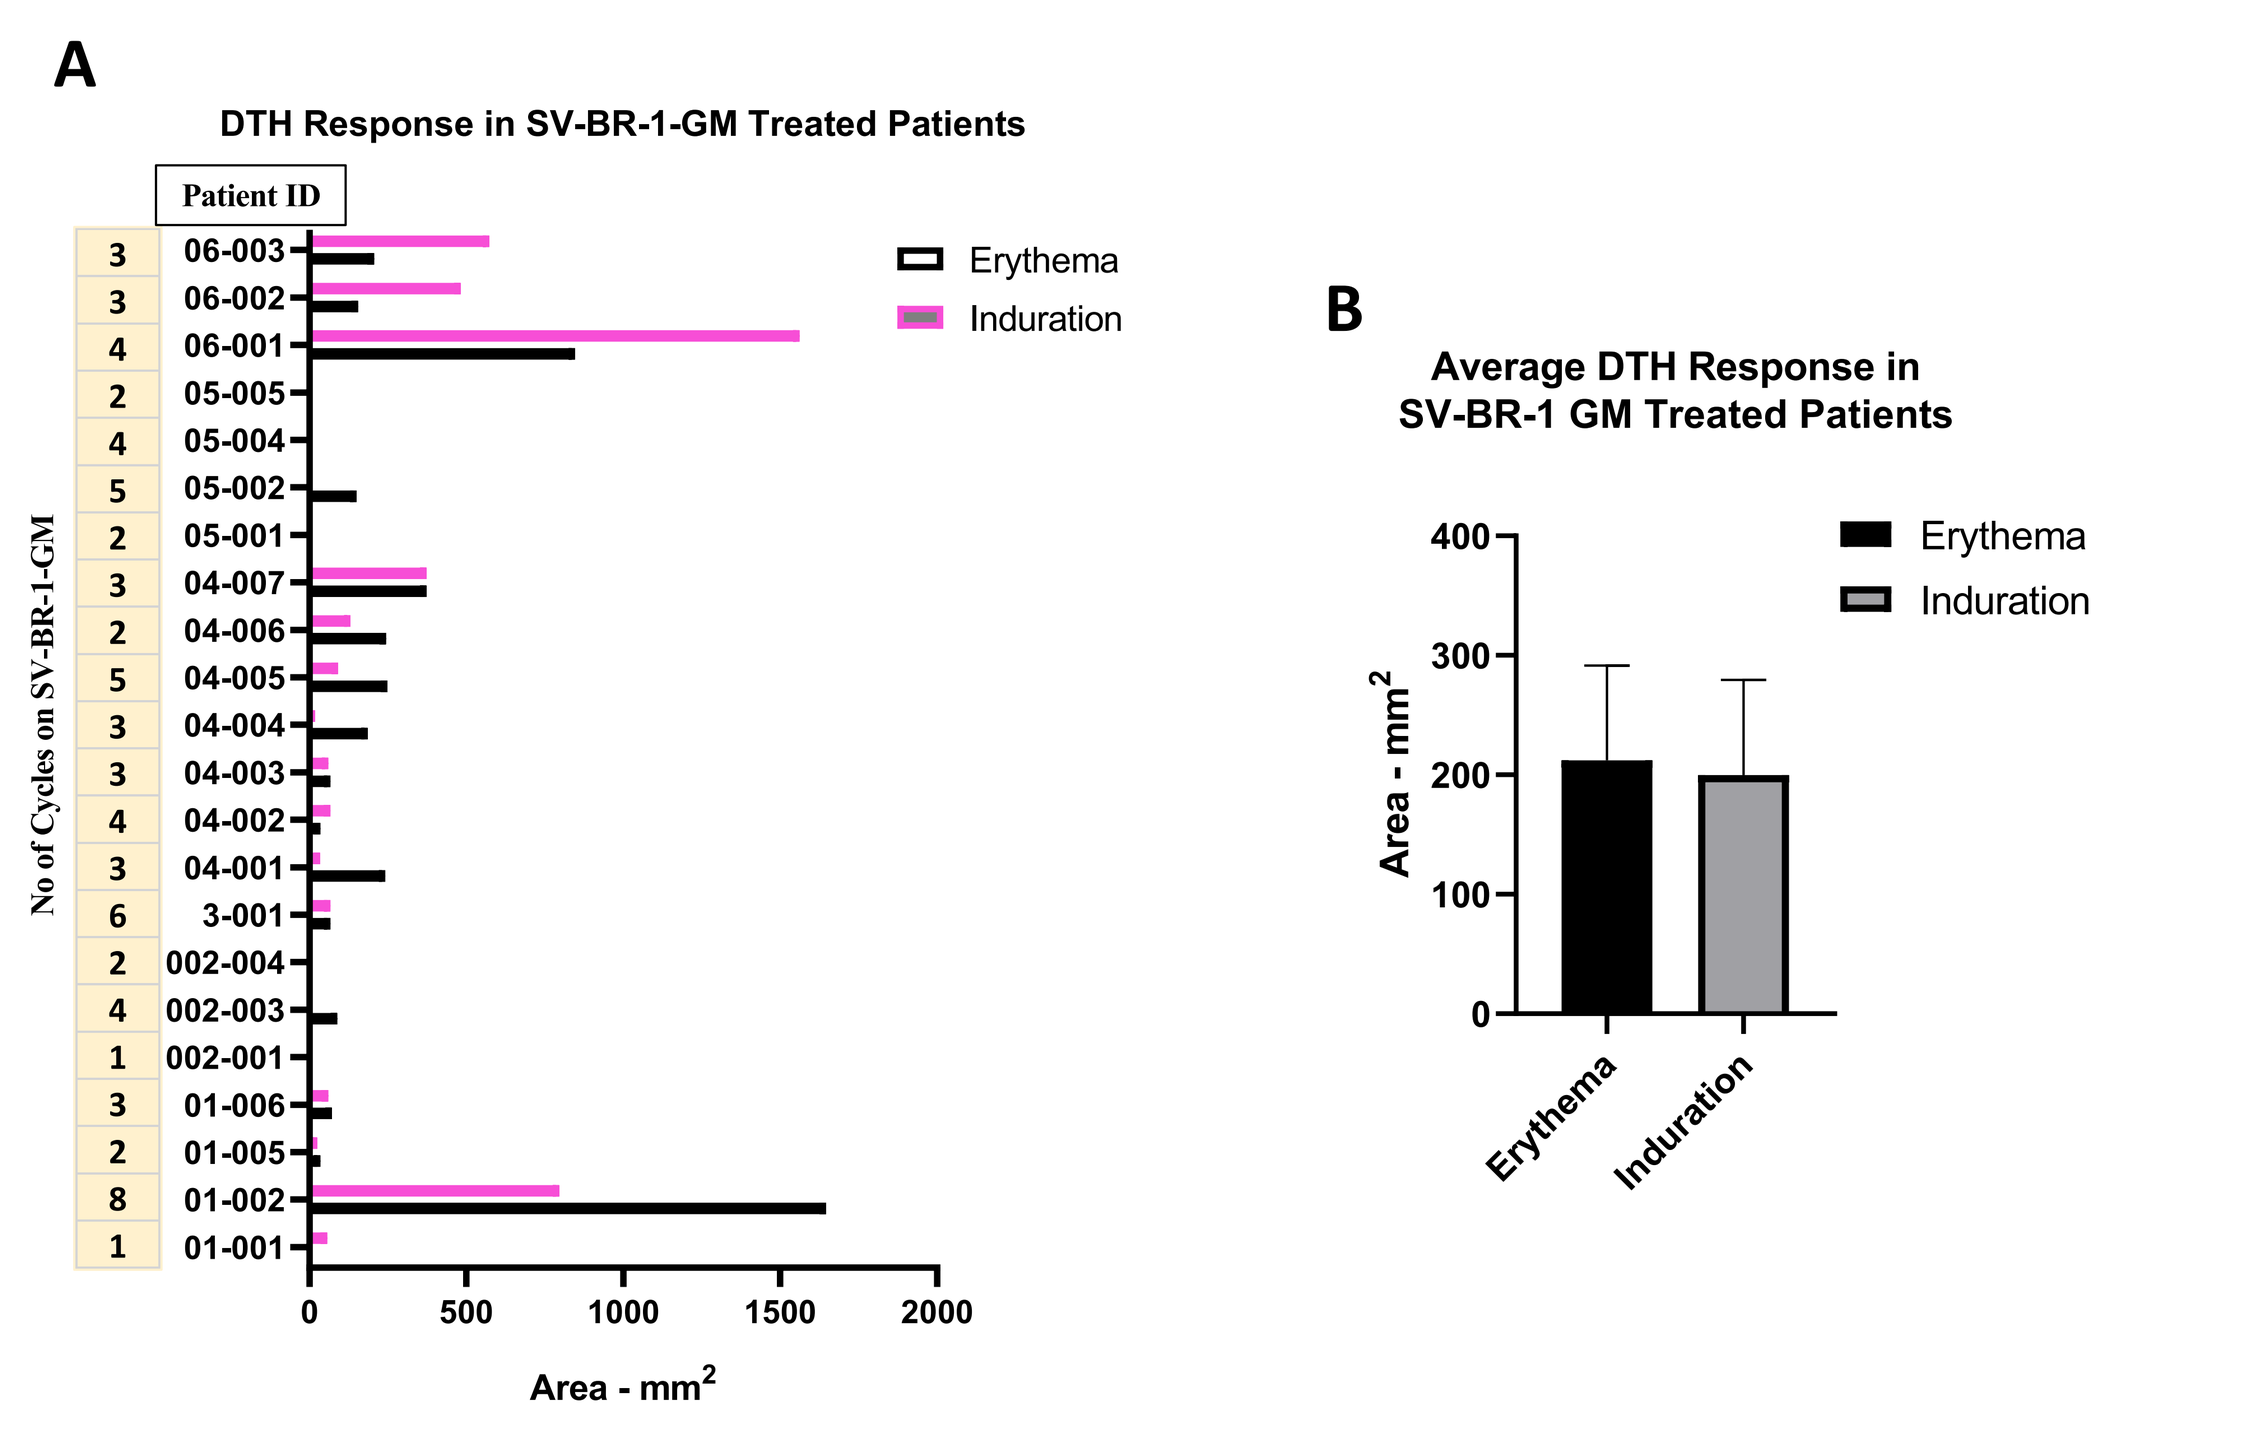
A
DTH Response in SV-BR-1-GM Treated Patients
Patient ID
No of Cycles on SV-BR-1-GM
Erythema
Induration
Area - mm²
B
Average DTH Response in SV-BR-1 GM Treated Patients
Area - mm²
Erythema
Induration

Figure 2: DTH response
Briefly, SV-BR-1-GM was injected intra-dermally in 4 sites in the upper back and thighs. 48 hours (±24 hours) later, these sites were assessed for erythema and induration. The Area (**mm²**) for Erythema and Induration are the highest response for each patient in their corresponding cycle's, average of all the cycles. Number of cycles each patient undergone are represented next to patient ID.
Conclusion: Most patients with follow-up information develop DTH to SV-BR-1-GM, in spite of anergy to test antigens (Candida) in some patients. The most robust response was seen in a patient with regression of multiple pulmonary metastases (01-002).

RESULTS

Anti-SV-BR-1 Antibody (IgG) titers in patient sera

Antibody titers are important indicators of immune response, Sera from SV-BR-1-GM treated patients were assessed for anti-SV-BR-1 antibodies.

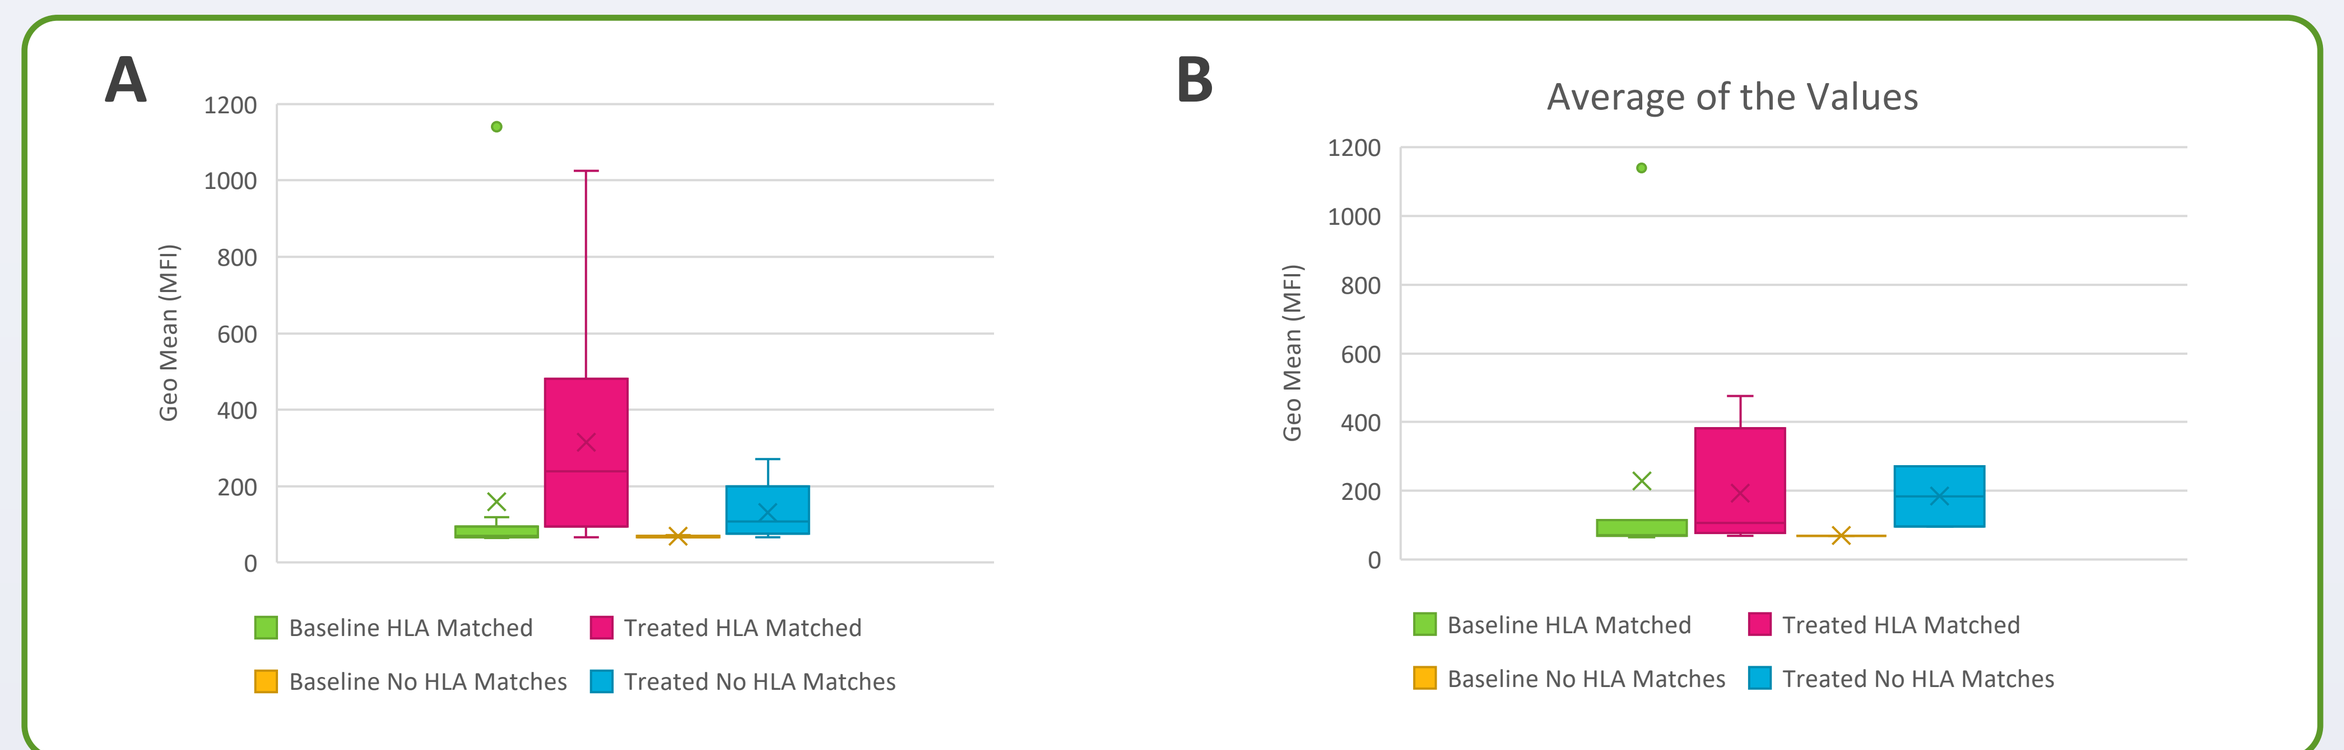
A
Geo Mean (MFI)
Baseline HLA Matched
Treated HLA Matched
Baseline No HLA Matches
Treated No HLA Matches
B
Average of the Values

Figure 3. Anti-SV-BR-1 antibody titers in patient sera.
SV-BR-1 cells were incubated with 1:10 diluted patient sera then stained with fluorescently labeled anti-human IgG and analyzed by flow cytometry. **A.** Anti-SV-BR-1 antibodies in all patient's sera samples. **B.** Data as in A. but samples segregated either as HLA matched (≥ 1 allele) or non-HLA matched. Baseline: before treatment with first dose of SV-BR-1-GM.
Conclusion: The SV-BR-1-GM regimen is able to elicit antibodies to SV-BR-1, the parent cell line, in patients with and without HLA matches.

Circulating Cancer-Associated Macrophage-Like Cells (CAMLs)

CAMLs are giant macrophage-like cells associated with patient tumors and found in the circulation of cancer patients from a variety of cancer types. The presence of tumor markers in CAMLs suggests that CAMLs phagocytose tumor material (Adams et al., 2014). Reduction in CAML frequency following treatment may indicate a favorable prognosis. **Figure 4** indicates that subjects with at least 1 HLA allele match to SV-BR-1-GM tend to respond with reduction in their CAML numbers. Note that PD-L1 expression was seen on CAMLs in 21/23 patients.

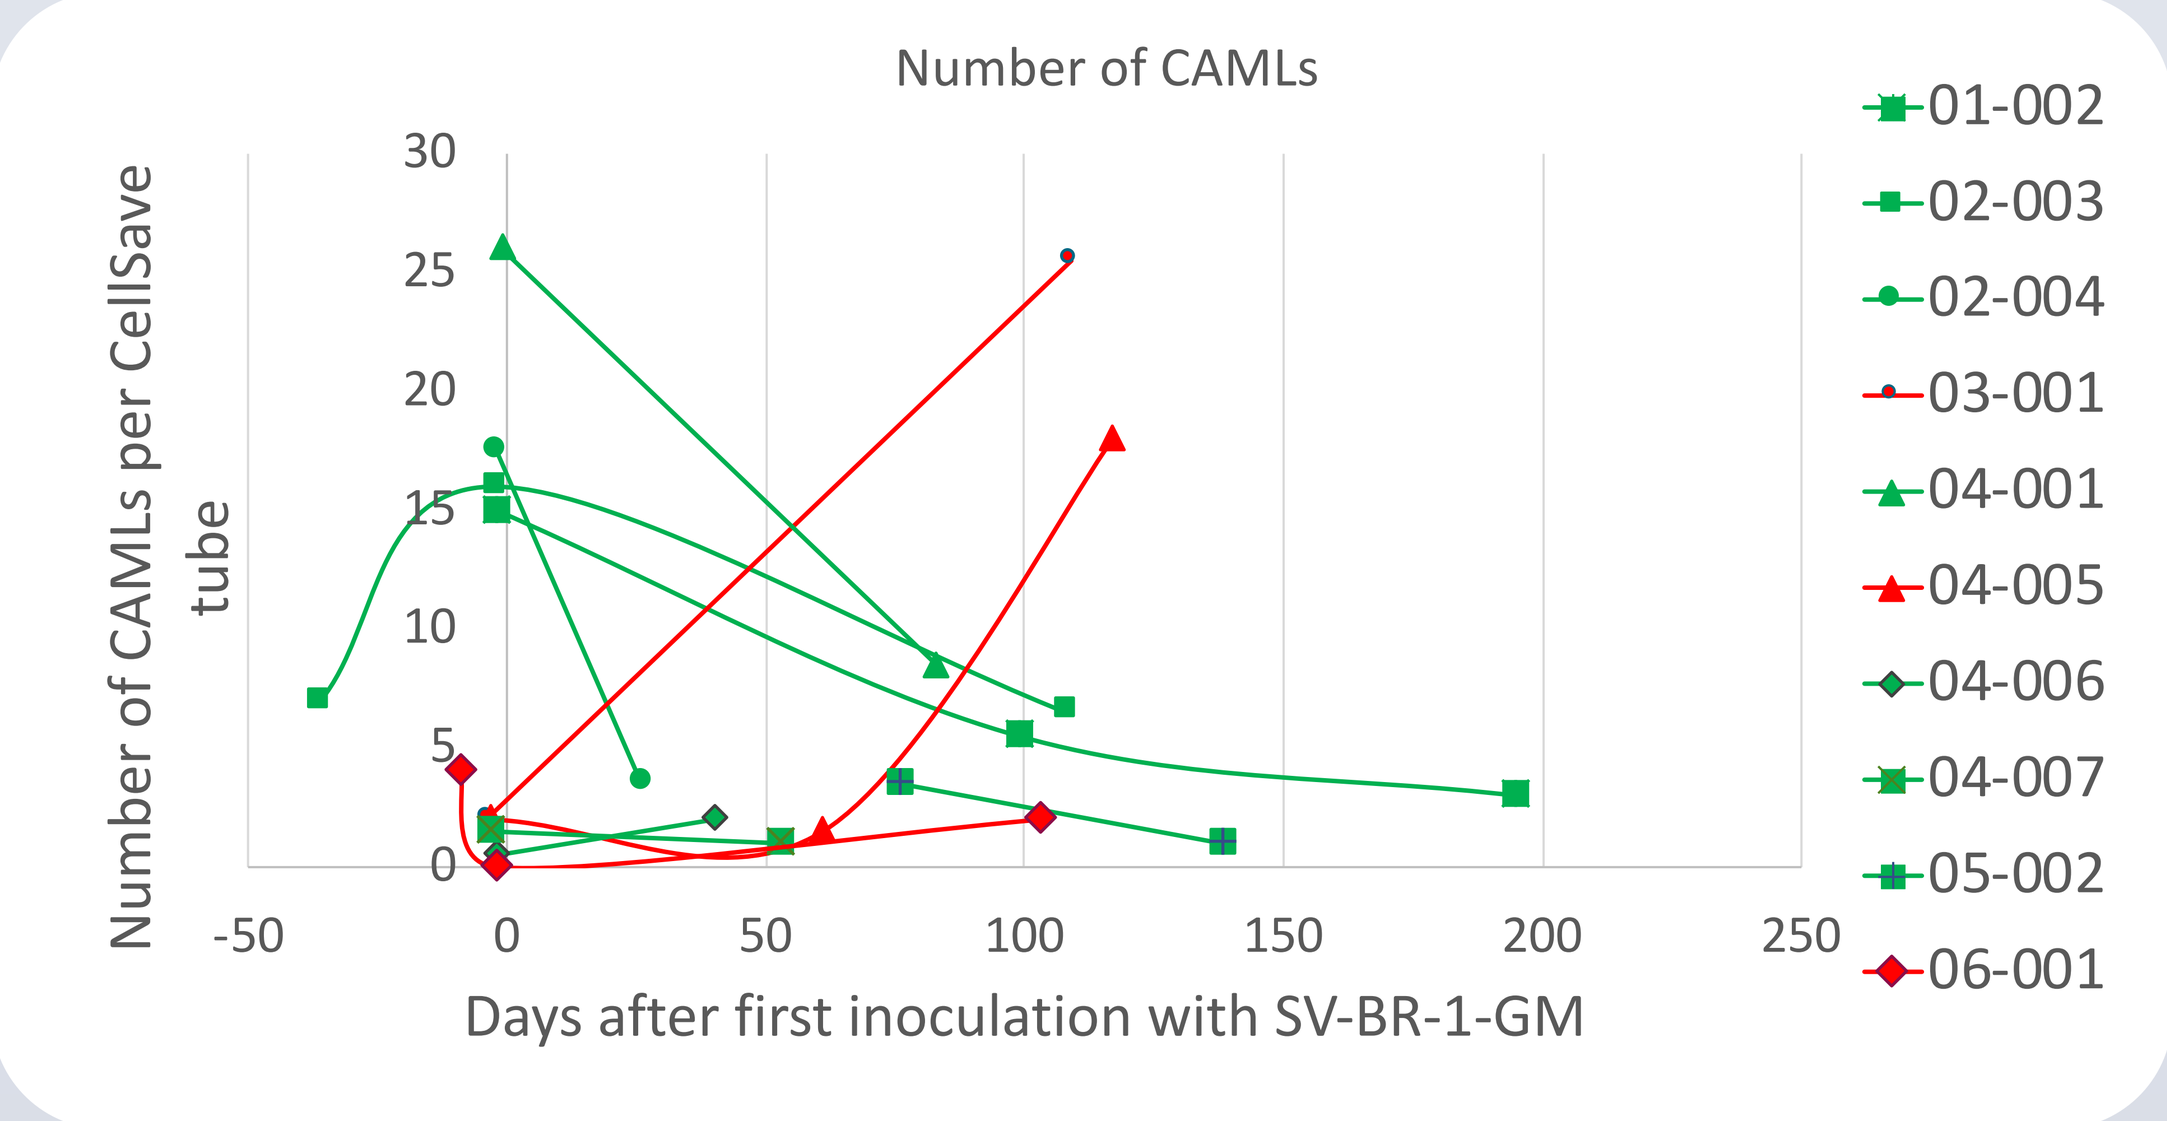
Number of CAMLs
Number of CAMLs per CellSave tube
Days after first inoculation with SV-BR-1-GM
01-002
02-003
02-004
03-001
04-001
04-005
04-006
04-007
05-002
06-001

Figure 4. CAMLs following SV-BR-1-GM inoculation. Reduction of CAML numbers in subjects with at least 1 HLA allele match to SV-BR-1-GM (green) compared to mismatching subjects (red). Subjects **01-002** and **05-002** experienced tumor regressions.
Conclusion: Reduction in CAMLs was seen especially in HLA-matched subjects, supporting our mechanism of action

Proposed Mechanism of Action of SV-BR-1-GM

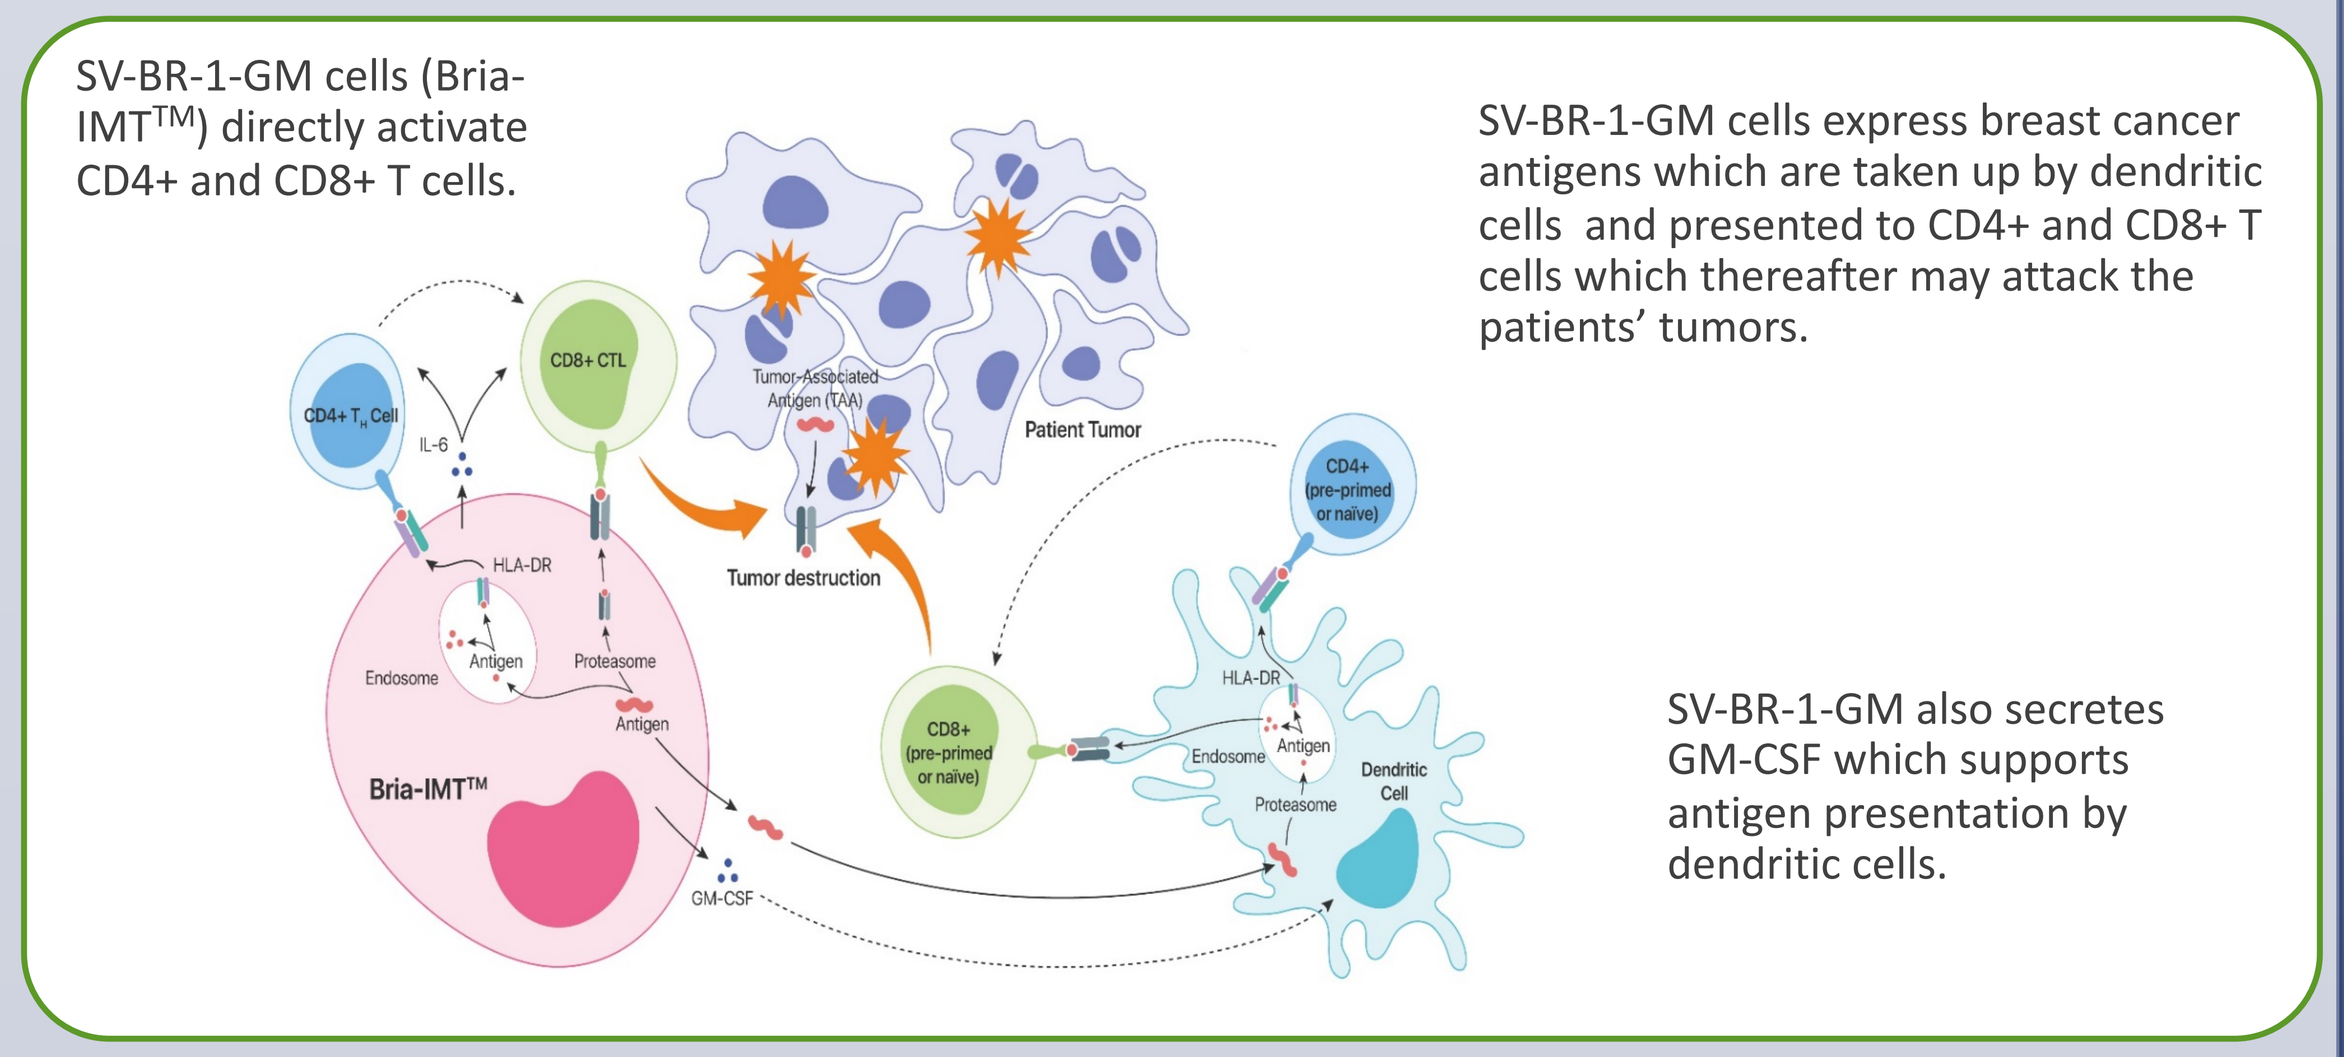
SV-BR-1-GM cells (Bria-IMT™) directly activate CD4+ and CD8+ T cells.
SV-BR-1-GM cells express breast cancer antigens which are taken up by dendritic cells and presented to CD4+ and CD8+ T cells which thereafter may attack the patients' tumors.
SV-BR-1-GM also secretes GM-CSF which supports antigen presentation by dendritic cells.
CD8+ CTL
Patient Tumor
Tumor destruction
Bria-IMT™
Dendritic Cell
GM-CSF

RESULTS – Study 2 Pembrolizumab Combination

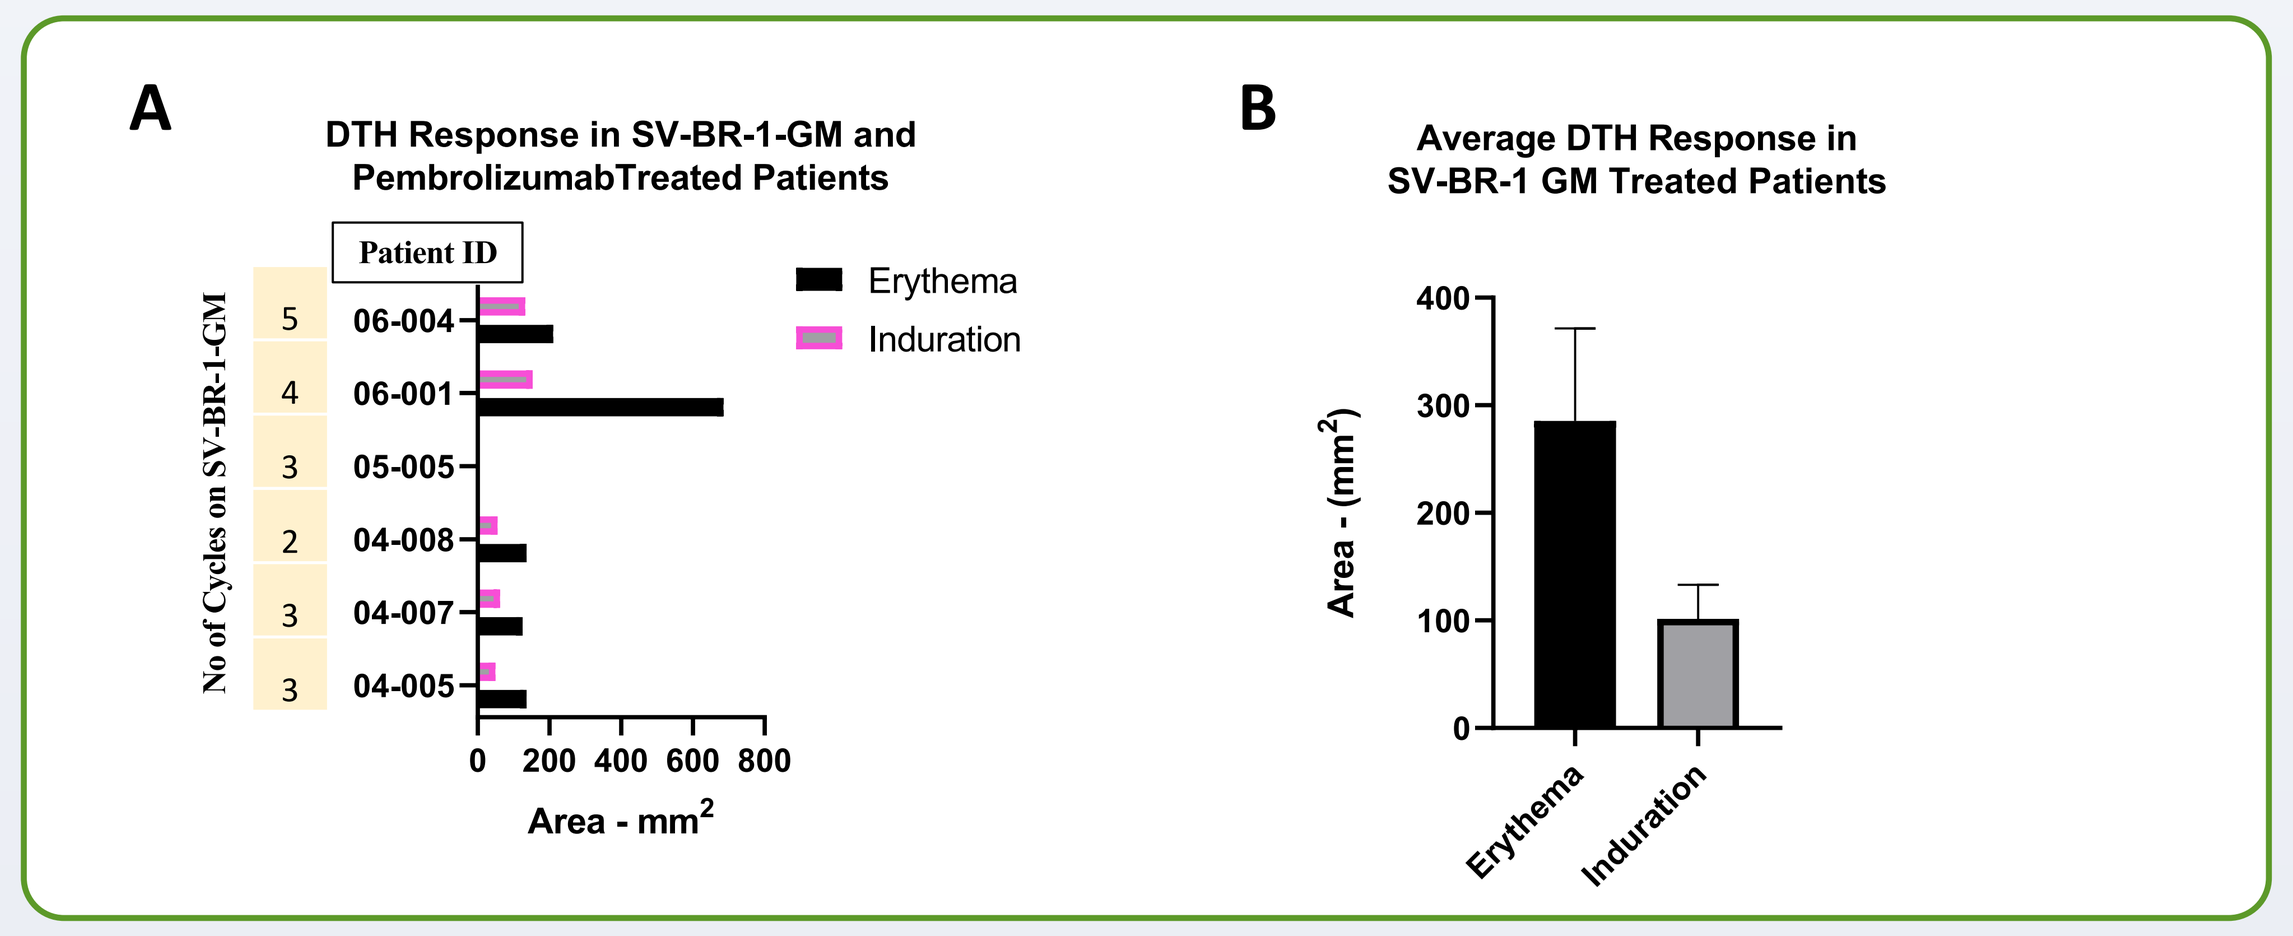
A
DTH Response in SV-BR-1-GM and PembrolizumabTreated Patients
Patient ID
No of Cycles on SV-BR-1-GM
Erythema
Induration
Area - mm²
B
Average DTH Response in SV-BR-1 GM Treated Patients
Area - (mm²)
Erythema
Induration

Figure 5: DTH response
Briefly, SV-BR-1-GM was injected intra-dermally in 4 sites in the upper back and thighs. 48 hours (±24 hours) later, these sites were assessed for erythema and induration. The Area (**mm²**) for Erythema and Induration are the highest response for each patient in their corresponding cycle's, average of all the cycles. Number of cycles each patient undergone are represented next to patient ID.
Conclusion: Most patients develop DTH to SV-BR-1-GM, in spite of anergy to test antigens (Candida) in some patients.

CONCLUSIONS

- **The SV-BR-1-GM regimen appears safe and well tolerated**
- **Evidence of tumor regression was seen primarily in patients who match SV-BR-1-GM at 1 or more HLA loci**
- **Delayed Type Hypersensitivity to SV-BR-1 is present in most patients after several cycles of treatment in both monotherapy and combination therapy patients and the most robust response was seen in a patient with regression of multiple pulmonary metastases**
- **Heightened expression of serum antibodies towards SV-BR-1-GM were elicited by the SV-BR-1-GM regimen in a preliminary analysis**
- **Reduction in CAMLs was seen especially in HLA-matched subjects, supporting our mechanism of action.**

REFERENCES

- Adams et al., Proc Natl Acad Sci U S A. 2014 Mar 4;111(9):3514-9
- Boegel et al., Genome Med. 2012 Dec 22;4(12):102
- Lacher et al., Front Immunol. 2018 May 15;9:776
- Wiseman and Kharazi; Breast J. 2006;12(5):475-80

ACKNOWLEDGEMENTS

We are grateful to Gerhard Bauer, Brian Fury, Emily Lynn Fledderman, Tye Daniel Petrie, Dane P. Coleal-Bergum, and Nicholas Perotti from the UC Davis GMP facility (Sacramento, CA) and to Don Healey and Lauren Collison from KBI Biopharma, Inc. for their SV-BR-1-GM manufacturing efforts. Creatv MicroTech for CAMLs measurements.

RESEARCH POSTER PRESENTATION DESIGN © 2015
www.PosterPresentations.com