Overall Survival following treatment with a modified whole tumor cell targeted immunotherapy in patients with advanced breast cancer

William V. Williams[1], Shaker R. Dakhil[2], Carmen Calfa[3], Jarrod P. Holmes[4], Saveri Bhattacharya[5], Jason Lukas[6], Elizabeth Tan-Chiu[7], George E. Peoples[8], Vivek Sunkari[1], Markus D. Lacher[1], and Charles L. Wiseman[1]

[1]BriaCell Therapeutics Corporation, Berkeley, CA; [2]Cancer Center of Kansas, Wichita, KS; [3]University of Miami, Miami, FL; [4]Redwood Reg Medical Grp, Santa Rosa, CA; [5]University of Pennsylvania, Philadelphia, PA; [6]The Everett Clinic, Everett, WA; [7]Florida Cancer Specialists and Research Institute, Parkland, FL; [8]Cancer Insight LLC, San Antonio, TX

San Antonio Breast Cancer Symposium – December 7-10, 2021

ABSTRACT

Background: SV-BR-1-GM is a GM-CSF secreting breast cancer cell line derived from a Grade II (moderately differentiated) breast tumor that also expresses HLA class I & II antigens and is able to function as an antigen-presenting cell. Irradiated SV-BR-1-GM is used in a regimen including pre-dose low-dose cyclophosphamide and post-dose injection of IFNα2b into the inoculation sites. The SV-BR-1-GM regimen has been used alone ("monotherapy", ClinicalTrials.gov NCT03066947 – study completed) and in combination with checkpoint inhibitors ("combination", ClinicalTrials.gov NCT03328026 – study ongoing). Here we report survival data for patients with advanced metastatic breast cancer (aMBC) treated with the SV-BR-1-GM regimen.

Methods: 27 patients with refractory aMBC were treated with the SV-BR-1-GM regimen as monotherapy (cycles every 2 weeks x3 and then monthly). The combination study uses the SV-BR-1-GM regimen with PD-1 inhibitors (PD-1i) pembrolizumab or retifanlimab with cycles every 3 weeks (12 patients dosed to date). Here we report progression free survival (PFS) and overall survival (OS) for patients where that data was collected.

Results: A total of 35 patients received the SV-BR-1-GM regimen. The SV-BR-1-GM regimen alone (monotherapy) was given to 27 and 12 received the regimen with a PD-1i checkpoint inhibitor (combination therapy): 4 subjects crossed over from monotherapy. Patients had been heavily pre-treated, median prior regimens = 5. Most patients were estrogen receptor and/or progesterone receptor positive, 18% were Her2/neu positive and 33% were triple negative. The treatment was generally safe and well tolerated. The disease control rate was 30% for the SV-BR-1-GM regimen alone and 33% for the combination with a PD-1i. Several patients had objective complete regression of selected metastases. Median progression free survival was 2.8 months for the SV-BR-1-GM regimen alone and 4.2 months for the PD-1i combination. Median overall survival was 7.0 months for the SV-BR-1-GM regimen alone (data available on 9 patients), and 12.0 months for the PD-1i combination (data available on 7 patients).

Conclusions: The median OS compares favorably with published data regarding survival in third line trials (Kazmi *Breast Cancer Res Treat*. 2020 Aug 17). The protracted OS seen in some subjects suggests some patient subpopulations are more likely to derive clinical benefit. The SV-BR-1-GM regimen alone or in combination with a PD-1i, when administered to heavily pre-treated patients with aMBC, may have elicited effective immune responses in some patients.

Table

	Patients by Study		
Characteristic	SV-BR-1-GM Regimen Alone (n=27)	SV-BR-1-GM Regimen + PD-1i (n=12)	All Patients* (n=35)
Age	60 ± 10	63 ± 10	60 ± 10
Mean Prior Systemic Regimens	5 (range 0-12)	6 (range 1-10)	5 (range 0-12)
% ER/PR +	52%	75%	58%
% Her2/neu +	15%	17%	18%
% Triple Negative	36%	25%	33%
Delayed-type Hypersensitivity	81%	91%	82%
Disease Control Rate	30%	33%	29%
Median (Range) Progression Free Survival (months)	2.8 (0.4-7.4) (n=27)	4.2 (0.8-9.4) (n=11)	2.8 (0.4-9.4) (n=34)
Median (Range) Overall Survival (months)	7.0 (1-41) (n=9)	12.0 (5.1-21.4) (n=7)	10.2 (1-41) (n=14)

- Note that 4 patients crossed over from the monotherapy study to the combination therapy study.

BACKGROUND AND OBJECTIVES

- SV-BR-1-GM is a breast cancer cell line with features of antigen-presenting cells including expression of HLA class II molecules (Lacher et al., Front Immunol. 2018 May 15;9:776)
- SV-BR-1-GM was derived from a Grade II (moderately differentiated) breast cancer biopsy tumor.
- SV-BR-1-GM was used in 3 clinical studies: In both studies the SV-BR-1-GM regimen consisted of low dose cyclophosphamide to reduce immune suppression (300 mg/m^2 2-3 days prior to inoculation); 20-40 million irradiated SV-BR-1-GM cells intradermally split into 4 sites; and interferon-α2b (10,000 IU x 4) into the inoculation sites ~2 & ~4 days later with cycles every 2 weeks x3 then monthly. For combination therapy, pembrolizumab (200 mg IV) or retifanlimab (375 mg IV) was given in combination with the regimen from the Monotherapy study with cycles every 3 weeks

BACKGROUND AND OBJECTIVES (continued)

- **Monotherapy Study 1 (SVMC #01-026):** This study included 3 patients with metastatic breast cancer who had failed a median of 2 prior regimens and one patient with HER2+ ovarian cancer who had failed 6 prior regimens.
- **Monotherapy Study 2 (WRI-GEV-007):** This study included 23 patients with metastatic breast cancer who had failed a median of 5 (1-13) prior regimens.
- **Combination Therapy Study (BRI-ROL-001):** This study included 12 subjects to date with metastatic breast cancer who had failed a median of 6 (1-13) prior regimens
- We have observed that clinical benefit appears more likely in patients who match SV-BR-1-GM at one or more HLA alleles.
- We also have observed that SV-BR-1-GM, which was derived from a Grade II metastatic breast cancer tumor, has a genetic fingerprint similar to that of other breast cancer cell lines derived from Grade I or Grade II tumors. Clinical benefit also appears better in patients with Grade I or II tumors.
- Here we evaluate survival data based on study, HLA matching and tumor grade.

RESULTS

Patient Characteristics

Monotherapy Study 1	No HLA Allele Matches (n=2)	1+ HLA Allele Matches (n=2)	2+ HLA Allele Matches (n=1)	All Patients (n=4)
Ages	60, 72	58, 72	58	58, 60, 72, 72
Prior Regimens	?, 6	2, 2	2	2, 2, 6, ?
% ER/PR +	100% (1/1)	100%	100%	100%
% Her2/neu +	100%	100%	100%	100%
% Triple Negative	0%	0%	0%	0%
Disease Control*	0%	50%	100%	25%
Grade I/II	NA	1/1	1/1	1/2

Monotherapy Study 2	No HLA Allele Matches (n=6)	1+ HLA Allele Matches (n=17)	2+ HLA Allele Matches (n=5)	All Patients (n=23)
Age	55 ± 14	60 ± 8	68 ± 7	59 ± 10
Prior Regimens	5.5 (range 2-13)	4 (range 0-10)	3.5 (range 3-7)	5 (range 1-13)
% ER/PR +	67%	47%	67%	52%
% Her2/neu +	17%	20%	33%	19%
% Triple Negative	33%	40%	0%	38%
Disease Control*	29%	30%	67%	30%
Grade I/II	2/2	50% (2/4)	100% (2/2)	67% (4/6)

Combination Therapy Study	No HLA Allele Matches (n=4)	1+ HLA Allele Matches (n=8)	2+ HLA Allele Matches (n=6)	All Patients# (n=12)
Age	61 ± 11	60 ± 10	60 ± 11	61 ± 10
Prior Regimens	7 (range 2-9)	5 (range 1-10)	4 (range 1-10)	6 (range 1-10)
% ER/PR +	75%	75%	67%	75%
% Her2/neu +	0%	25%	17%	17%
% Triple Negative	25%	25%	33%	25%
Disease Control*	25%	38%	33%	33%
Grade I/II	1/1	2/3	1/2	3/4

*Disease Control includes total of 8 with stable disease (SD) and 2 with partial responses (PR). Both PRs matched SV-BR-1-GM at 2 HLA loci and both had Grade II disease

#Four patients crossed over from Monotherapy Study 2 to the Combination Therapy Study

RESULTS CONTINUED

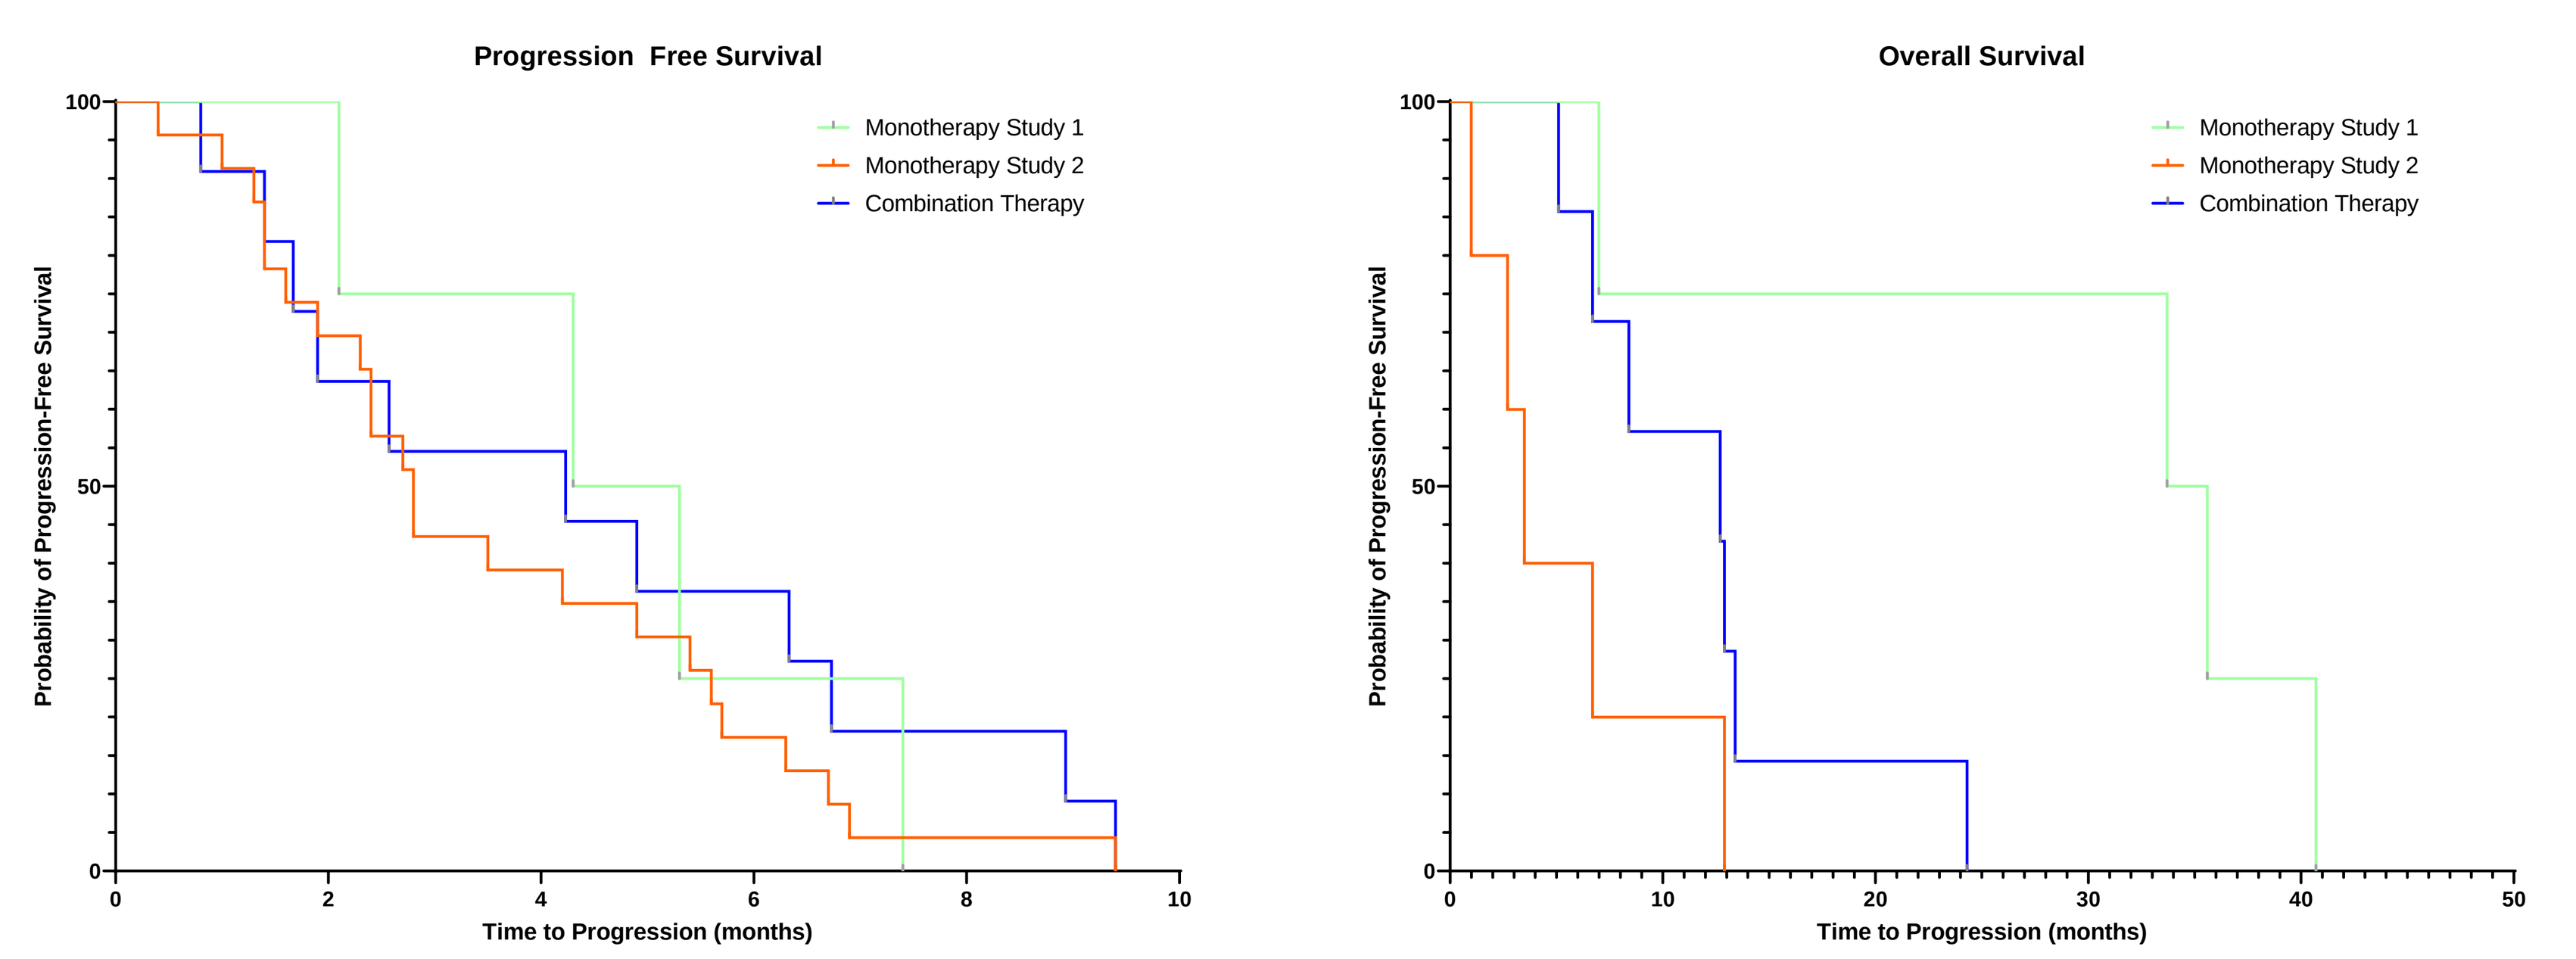
Progression Free Survival
Overall Survival
Monotherapy Study 1
Monotherapy Study 2
Combination Therapy
Probability of Progression-Free Survival
Time to Progression (months)

Figure 1. Progression Free Survival (PFS) and Overall Survival (OS) by Study. PFS is shown on the left and OS on the right.

Median PFS was 4.78 months for monotherapy study 1 (n=4), 2.80 months for monotherapy study 2 (n=23) and 4.23 months for the combination therapy study (n=11). Median OS was 34.65 months for monotherapy study 1 (n=4), 7.0 months for monotherapy study 2 (n=5) and for the combination therapy study was 12.7 months (n=7). Note that the patients in monotherapy Study 1 were in general less heavily pre-treated.

Conclusion: PFS and OS in this group of heavily pre-treated metastatic breast cancer patients appears better for the combination therapy group compared with the monotherapy group.

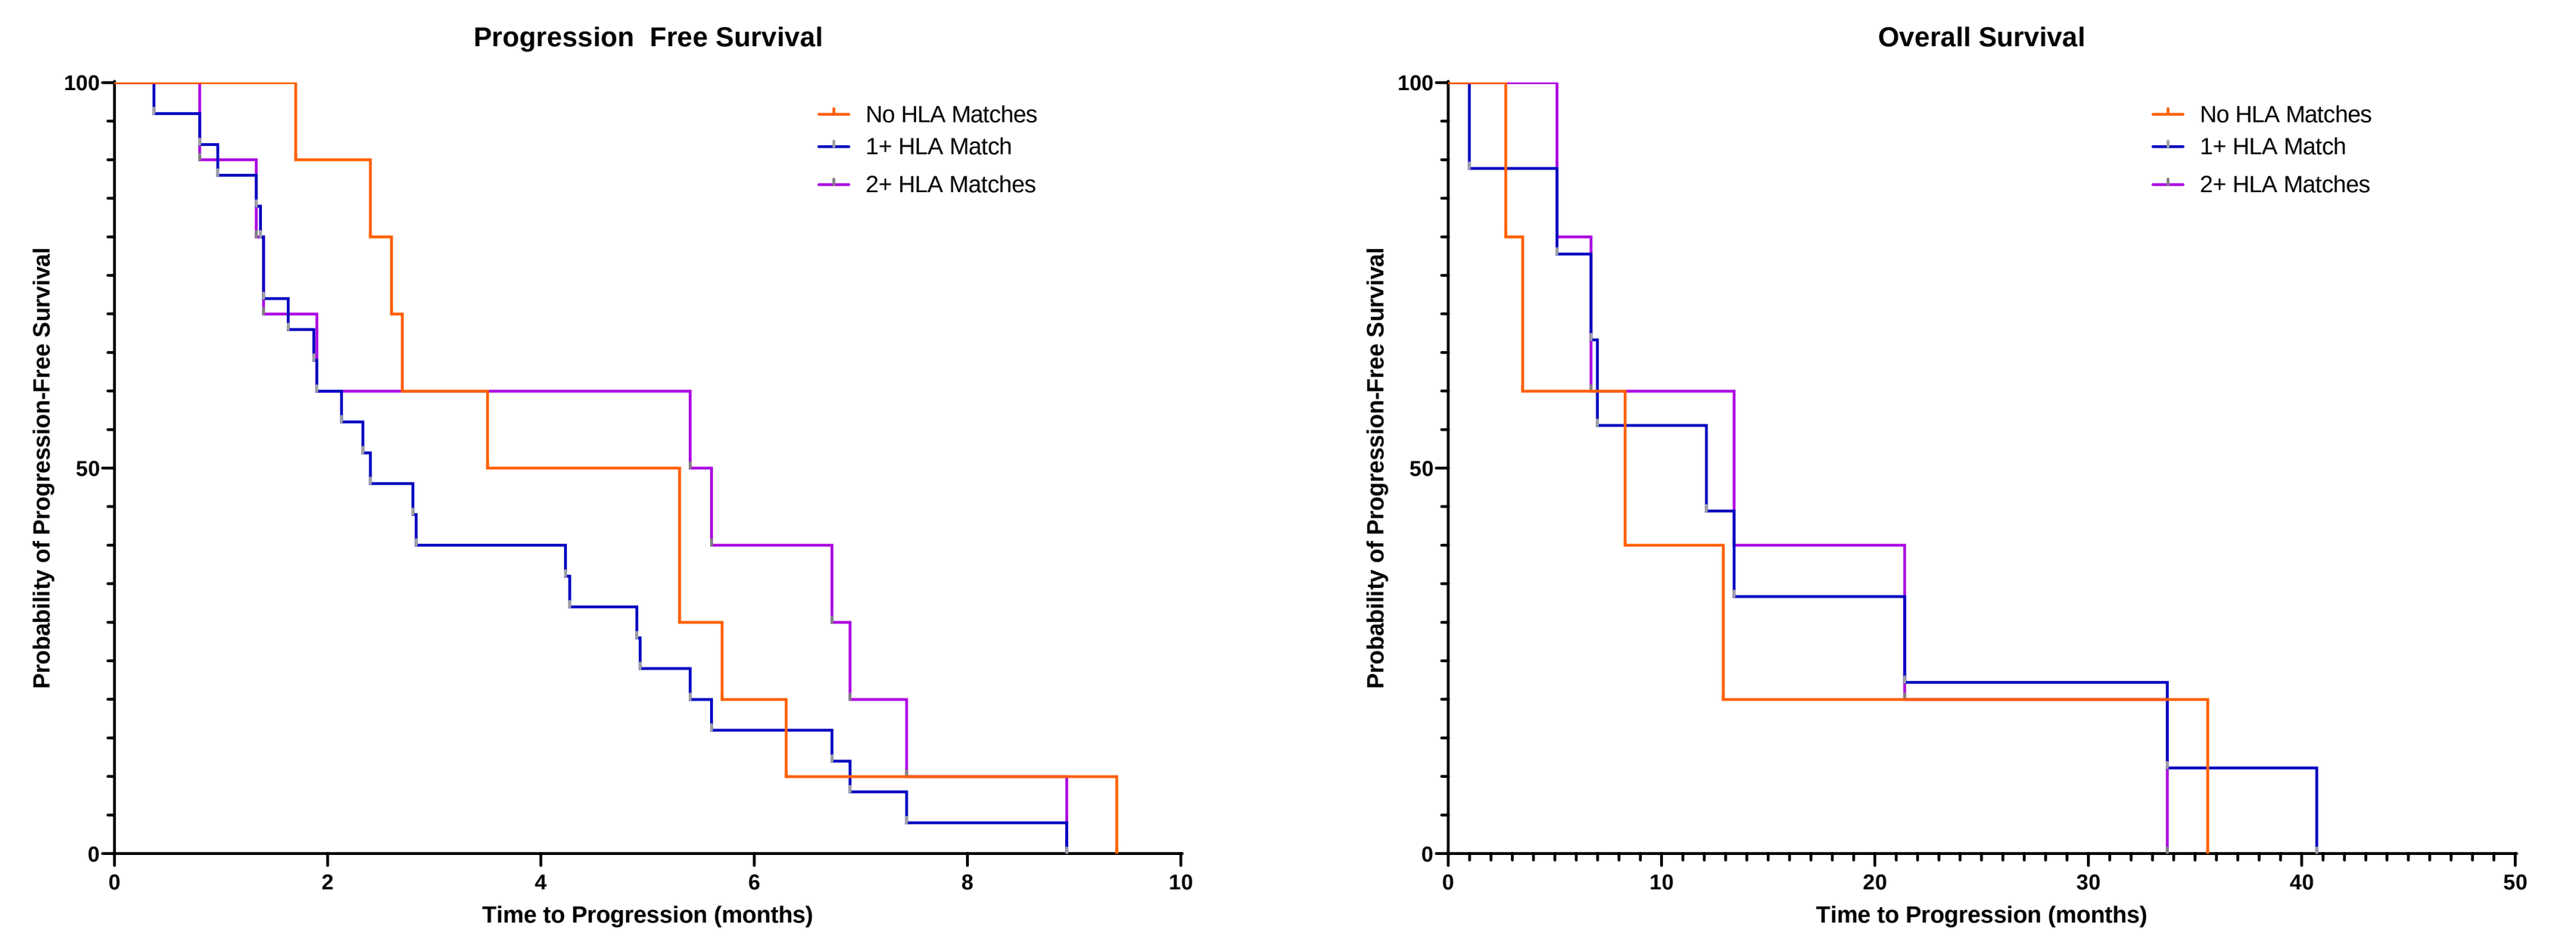
Progression Free Survival
Overall Survival
No HLA Matches
1+ HLA Match
2+ HLA Matches
Probability of Progression-Free Survival
Time to Progression (months)

Figure 2. PFS and OS by HLA Matching. PFS is shown on the left and OS on the right.

Median PFS was 3.1 months for those with no HLA matches (n=9), 2.4 months for those with 1+ HLA matches (n=25) and 5.5 months for those with 2+ HLA matches (n=11).

Median OS was 5.9 months for those with no HLA matches (n=4), 12.7 months for those with 1+ HLA matches (n=8) and 13.4 months for those with 2+ HLA matches (n=5)

Conclusion: PFS and OS in this group of heavily pre-treated metastatic breast cancer patients appears better for those 1+ and 2+ HLA matches with SV-BR-1-GM compared with those with no HLA matches.

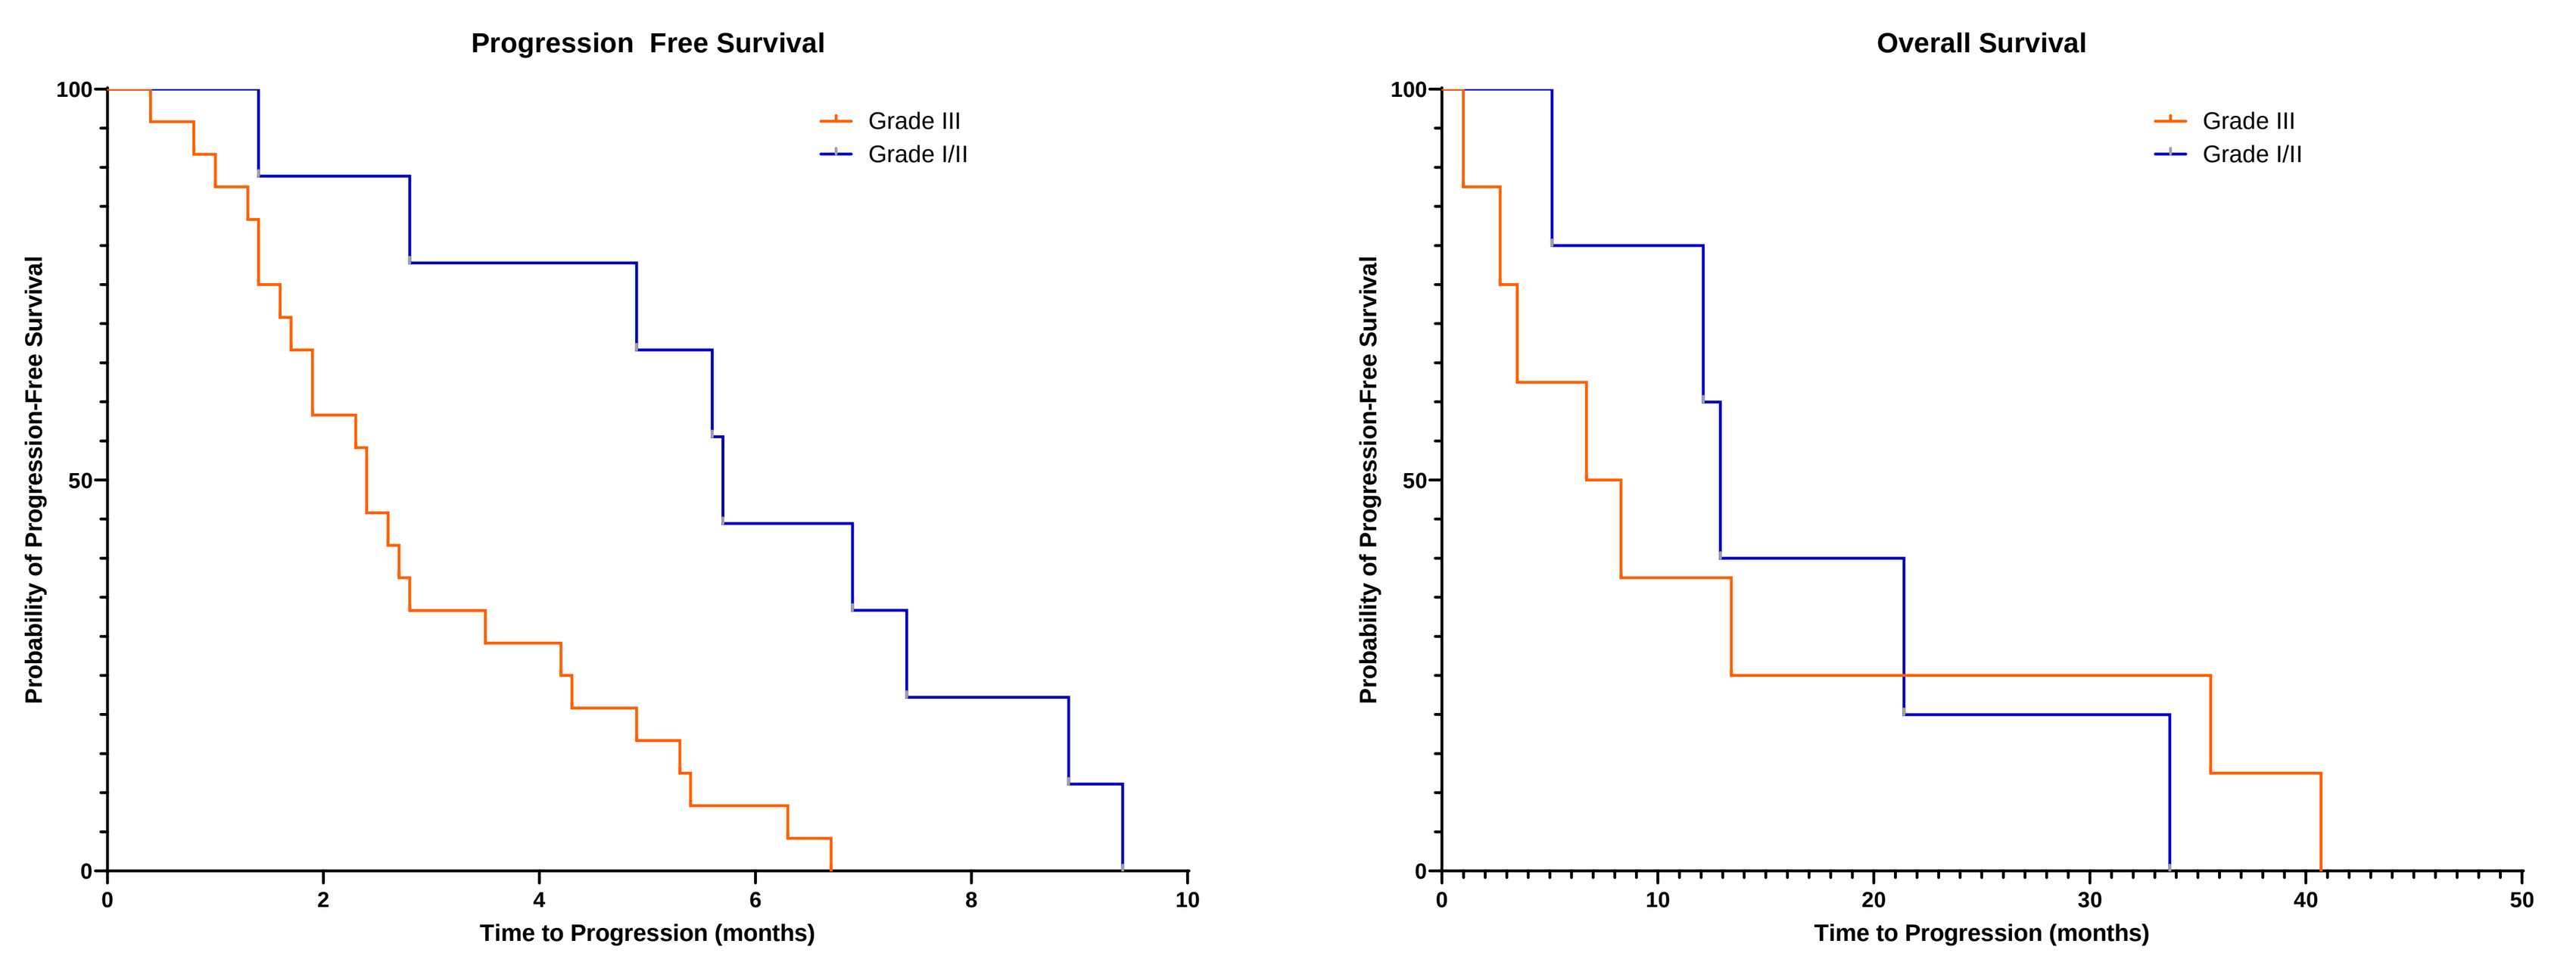
Progression Free Survival
Overall Survival
Grade III
Grade I/II
Probability of Progression-Free Survival
Time to Progression (months)

Figure 3. PFS and OS by Tumor Grade. PFS is shown on the left and OS on the right.

Median PFS was 2.4 months for those with Grade III tumors (n=23) and 5.7 months for those with Grade I/II tumors (n=10).

Median OS was 6.7 months for those with Grade III tumors (n=7) and 12.9 months for those with Grade I/II tumors (n=5).

Conclusion: PFS and OS in this group of heavily pre-treated metastatic breast cancer patients appears better for those with Grade I/II tumors compared with those with Grade III tumors.

Conclusions and Hypotheses

- **Thirty-four breast cancer and one ovarian cancer patients have been treated with the SV-BR-1-GM regimen alone or in combination with a PD-1 inhibitor.**
- **These patients were very heavily pre-treated and had failed on average 5-6 prior regimens.**
- **The PFS and OS of combination therapy with a PD-1i appears superior to the monotherapy regimen when studies with similar patients are compared.**
- **PFS and OS appears better in patients who HLA match with SV-BR-1-GM and those with Grade I/II tumors.**
- **The ongoing combination therapy study will focus on patients most likely to benefit (those with at least 1 HLA match and those with Grade I or II tumors)**

References: Kazmi S et al, 2020 Breast Cancer Res Treat. 2020 Aug 17. doi: 10.1007/s10549-020-05867-0; Lacher MD et al, Front Immunol. 2018 May 15;9:776 – see for prior characterization of SV-BR-1-GM

This presentation is the intellectual property of the author/presenter. Contact them at info@briacell.com for permission to reprint and/or distribute.